PAGE meeting, 5-8 June, 2012, Venice – Italy

The impact of using the well-stirred liver model in PBPK modelling for high-clearance compounds

Ruben de Kanter, Shirin Bruderer, Jochen Zisowsky, Jasper Dingemanse, Andreas Krause, Alexander Treiber and Christopher Kohl
Drug Metabolism and Pharmacokinetics (RdK, CK, AT) and Clinical Pharmacology (JZ, JD, AK)
Actelion Pharmaceuticals Ltd. / Gewerbestrasse 16 / CH-4123 Allschwil / Switzerland. ruben.de-kanter@actelion.com

Aim: The well-stirred liver model is a widely used standard in physiologically-based pharmacokinetic (PBPK) modelling. This analysis aimed at improving the prediction of the food effect observed for a high-clearance oral drug, assumed to be caused by a change in hepatic blood flow due to the presence of food.

Introduction

One of many causes for the effect of food on changes in drug exposure is a change in blood flow through the liver. After oral dosing, absolute bioavailability (F) can be defined as the fraction of the oral dose that is absorbed and not extracted by the liver and/or gut. The hepatic extraction ratio, E, is defined as hepatic clearance (CL_h) divided by the liver blood flow (Q_h). A change in liver blood flow will therefore have an effect on hepatic extraction ratio ($E = CL/Q_h$) and thus on bioavailability (F = 1-E), if a drug is completely absorbed and only hepatic clearance takes place.
Three hepatic models are in wider use for modelling the hepatic clearance: well-stirred model, parallel tube model, and dispersion model. The dispersion model is a general model that includes the performance of both the well-stirred model and the tube model. The differences between these models have been studied and although in most cases the different models give similar clearance values, there are differences, especially for high clearance drugs [1] .
Both the well-stirred and the parallel tube models assume instant equilibrium between the hepatocyte and adjacent blood. The major difference between these two models is that the well-stirred model considers the distribution of a drug in the liver to be homogeneous, whereas the parallel tube model assumes a declining hepatic drug concentration along the direction of blood flow.

Aim: investigate the performance of the well-stirred and the parallel tube liver models and the PBPK software package GastroPlus™ to describe the effects of changed liver blood flow, due to the presence of food, on clearance, hepatic extraction ratio and bioavailability.

Methods

Calculations: It was assumed that only hepatic clearance (CL_h) takes place and that bioavailability (F) is defined by F = 1- E, where E is the hepatic extraction ratio CL_h/Q (Q_h is the hepatic blood flow)

The following equations were used, where CL_{int} is the intrinsic clearance and f_u is the unbound fraction in blood:

for the well-stirred model: $_h = \frac{_h \cdot \quad \cdot}{_h + \quad \cdot}$, for the parallel tube model: $_h = \ _h \ 1 - \quad - \frac{\cdot}{h}$

PBPK modelling: GastroPlus™ v.7 (Simulations Plus Inc.) was applied with the following assumptions: 72% absorption (predicted permeability, dose 500 mg), predicted volume of distribution of 283 L and a hepatic blood clearance of 12.5 mL/min/kg. Default human PBPK physiology was applied for the fasted state, and the liver blood flow was increased from 1.5 L/min to 2 L/min for the fed state [2].
It was assumed that no saturation takes place (i.e. concentrations << Km so that CL_{int} equals V_{max}/K_m)

Results – PBPK modelling

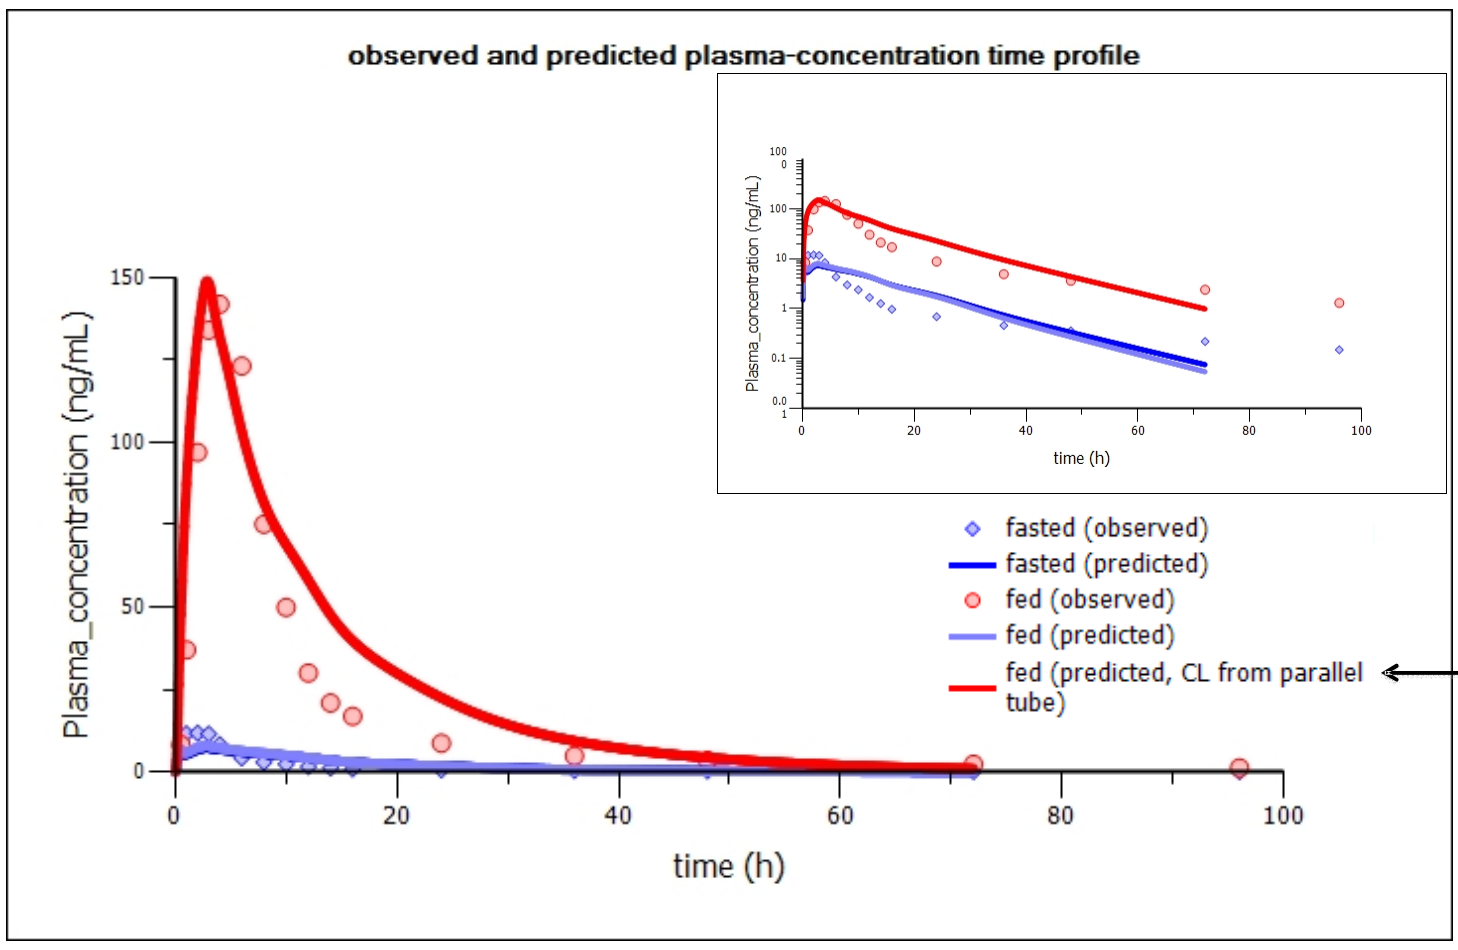
observed and predicted plasma-concentration time profile
Plasma_concentration (ng/mL)
time (h)
fasted (observed)
fasted (predicted)
fed (observed)
fed (predicted)
fed (predicted, CL from parallel tube)

•observed food effect: 15-fold increase in AUC
•predicted food effect (PBPK – well stirred model): no increase in AUC
•predicted food effect (PBPK – CL from parallel tube model): 15-fold increase in AUC

Discussion and Conclusion

A key objective for pre-clinical research is the prediction of human pharmacokinetics for compounds entering clinical studies. Often the entry into humans study is performed in fasted subjects and when a food effect on PK is suspected (for example based on animal studies), it is being studied early in clinical development using a standardized high-fat breakfast. PBPK modeling has been shown to be very useful in the prediction of food effects caused by the gastrointestinal physiology (pH, fluid volume, transit time) and drug dissolution and absorption [3]. Modeling can also be used to describe and understand the effect of food on changed liver blood flow, both after oral and iv administration [4].

We show here that the PBPK prediction of the food effect in for high-clearance drugs can be improved by a refinement of the liver model.

Results – liver model comparison

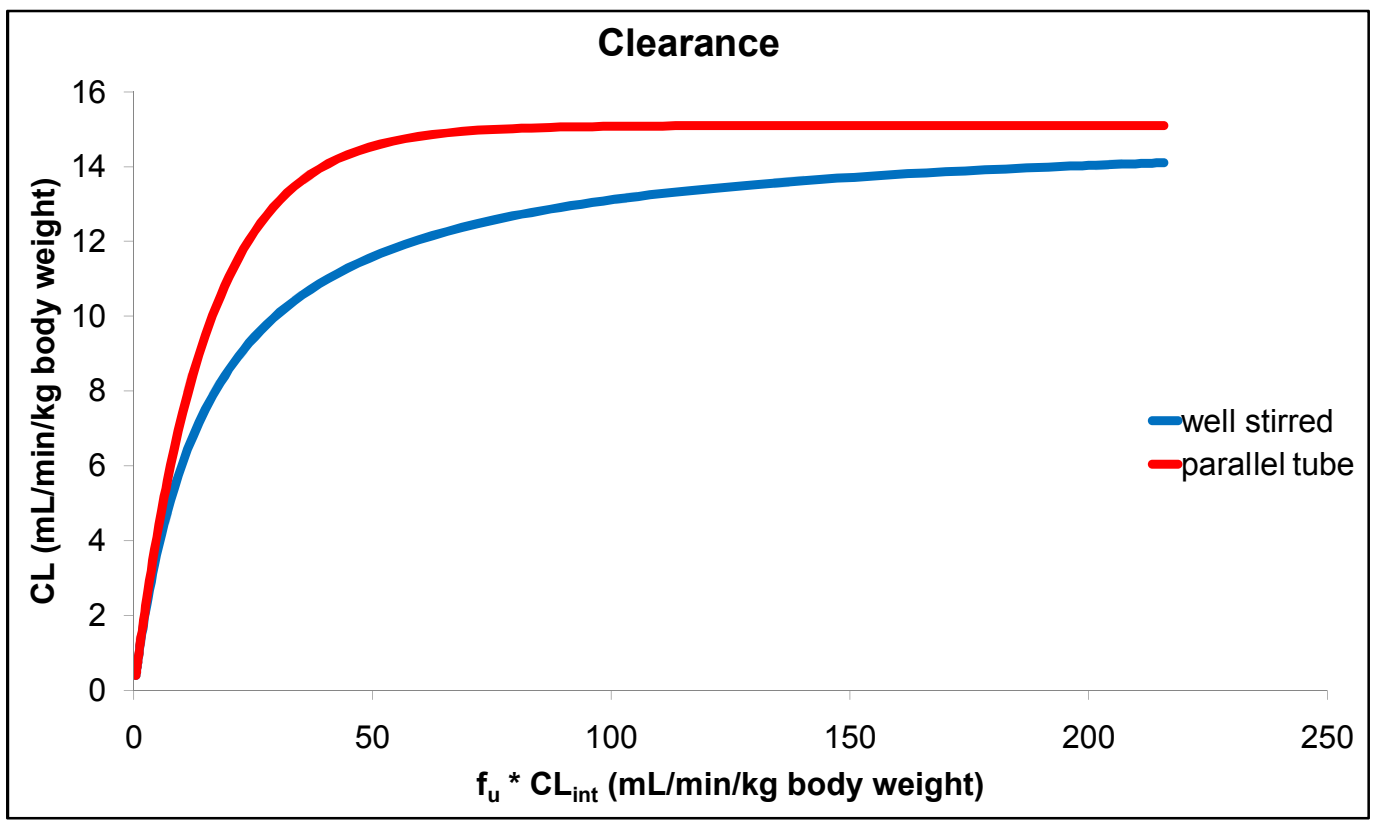
Clearance
CL (mL/min/kg body weight)
f_u * CL_int (mL/min/kg body weight)
well stirred
parallel tube

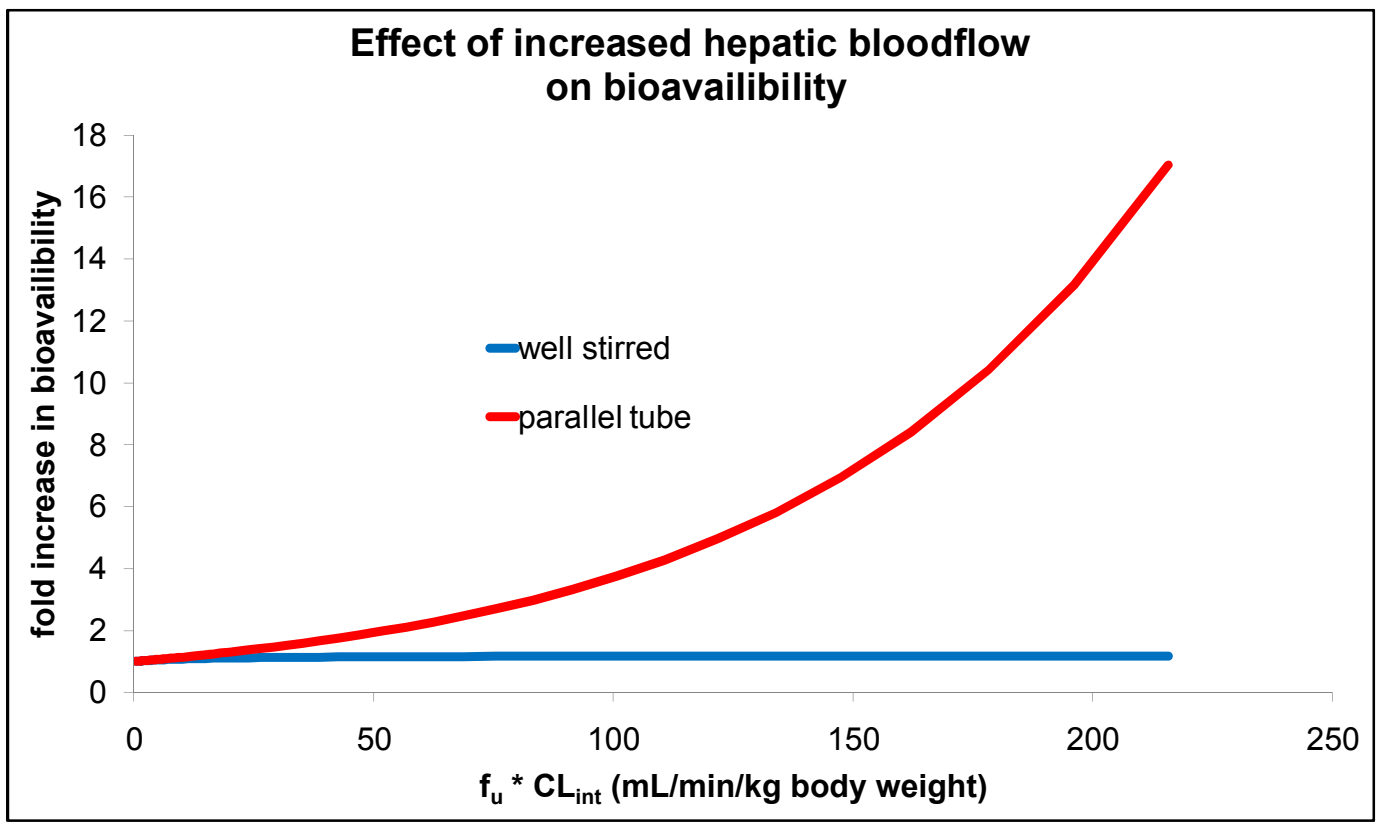
Effect of increased hepatic bloodflow on bioavailibility
fold increase in bioavailibility
f_u * CL_int (mL/min/kg body weight)
well stirred
parallel tube

well-stirred	f_u * CL_{int} (mL/min/kg)	Q_h (mL/min/kg)	CL_h (mL/min/kg)	hepatic extraction	bioavailability	fold increase
fasted	206	12.6	11.9	94%	6%	
fed	206	15.1	14.1	93%	7%	1.2
parallel tube	f_u * CL_{int} (mL/min/kg)	Q_h (mL/min/kg)	CL_h (mL/min/kg)	hepatic extraction	bioavailability	fold increase
fasted	206	12.6	12.6	99.99999%	0.00001%	
fed	206	15.1	15.1	99.99988%	0.00012%	15.0

References:
[1] Ito and Houston, Pharm Res 21, 785, 2004
[2] GastroPlus manual
[3] Parrott, Lukacova, Fraczkiewicz and Bolger, AAPS, 11, 45, 2009
[4] Nicolas, Krause, Gutierrez and Dingemanse, Br J Clin Pharmacol, 2012; Accepted Article.